Treatment Sequencing Patterns in Medicare-Enrolled Patients with Multiple Myeloma

Shuling Li, PhD,[1] Tanya Natwick, BA,[1] Akeem Yusuf, PhD,[2,3] Sarah Vidito, MD,[3] Khalid Mezzi, MD,[3] Winifred Werther, PhD[4]

[1]Chronic Disease Research Group, Minneapolis Medical Research Foundation, Minneapolis, MN; [2]University of Minnesota, Minneapolis, MN; [3]Amgen Inc., Thousand Oaks, CA; [4]Amgen Inc., South San Francisco, CA

INTRODUCTION

- Over the last decade, several novel therapies have been approved for multiple myeloma (MM) leading to significant improvement in the survival of MM patients[1]
- MM patients are often treated with multiple lines of therapy as relapse occurs[2]
- With the multiple options of therapeutic agents and novel combinations of regimens, the treatment for MM has become more complex[3]
- However, little is known about the treatment sequencing patterns for the patients with MM
- In this study, we described the use of drug regimens by lines of therapy and characterized treatment sequences in Medicare patients with MM

METHODS

- Data source: The Centers for Medicare & Medicaid Services (CMS) 100% Hematologic Cancer File (2007-2012)
- Inclusion criteria:
 - Diagnosed with MM between January 1, 2008 and December 31, 2011 based on a validated algorithm using a combination of ICD-9-CM diagnosis codes 203.0X and either diagnostic tests or treatment.[4] The diagnosis date was defined as the disease index date
 - Initiated a first-line therapy following the disease index date. The date of treatment initiation was identified as the first-line index date
 - Continuously enrolled in Medicare Part A, Part B, and Part D during the time period from 12 months before the disease index date to the first-line index date
 - Aged 18 years or older at the disease index date
- Exclusion criteria:
 - Received chemotherapy and/or radiotherapy in the 12 months before the disease index date
 - Had evidence of stem cell transplant in the 12 months before the disease index date
 - Received combination therapies not recommended in the National Comprehensive Cancer Network (NCCN) guidelines (Version 2.2014) for first-line therapy
- Line of therapy:
 - Patients who advanced to second-, third-, and fourth-line were identified if a 90-day gap in all treatments was observed or when a drug was added to a regimen >90 days after the line index date
 - To further identify patients who initiated multiple lines of therapy, we required that they be continuously enrolled in Medicare Parts A, B, and D from first-line initiation to the current-line initiation and that they receive monotherapy, doublets, or triplets regimens for the current line
- Drug regimens:
 - Identified using National Drug Code from Medicare Part D prescription drug event claims and Healthcare Common Procedure Coding System codes from Part B line items and Part A outpatient claims
 - Classified as monotherapy, doublets, and triplets based on the NCCN MM treatment guidelines; dexamethasone and prednisone were identified as steroids when defining regimens
- The study period was from the first-line index date to the earliest of death, disenrollment from Medicare Parts A, B, and D coverage, receipt of treatments not recommended in the NCCN MM guidelines, fourth-line end date, or December 31, 2012
- We described the distribution of type of drug regimens by lines of therapy, overall, and by age defined at MM index date, and characterized treatment sequencing patterns for patients who advanced to second-, third-, and fourth-line by type of drug regimens in the prior line of therapy, respectively

RESULTS

- Of 73,028 patients with MM diagnosis between January 1, 2008 and December 31, 2011, we identified 12,563 (17%) patients who met the study inclusion and exclusion criteria **(Figure 1)**
- Of the patients included for analyses, 5647 (45%), 2243 (18%), and 773 (6%) patients advanced to second-, third-, and fourth-line, respectively. **Table 1** shows demographics and clinical characteristics for patients included in each line
 - Of patients that initiated first-line therapy, mean (standard deviation) age, defined at the disease index date, was 74.8 (8.5) years; 53% were female; 78% were white
 - Older patients (aged ≥75 years) were less likely to advance to later lines compared with younger patients (e.g., from first-line to fourth-line, the percent of older patients decreased from 50% to 36%; while the percent of patients aged 65-74 years increased from 42% to 55% and the percent of patients aged 18-64 years remained stable at 9%

Figure 1. Multiple myeloma (MM) patient flow through the study inclusion criteria

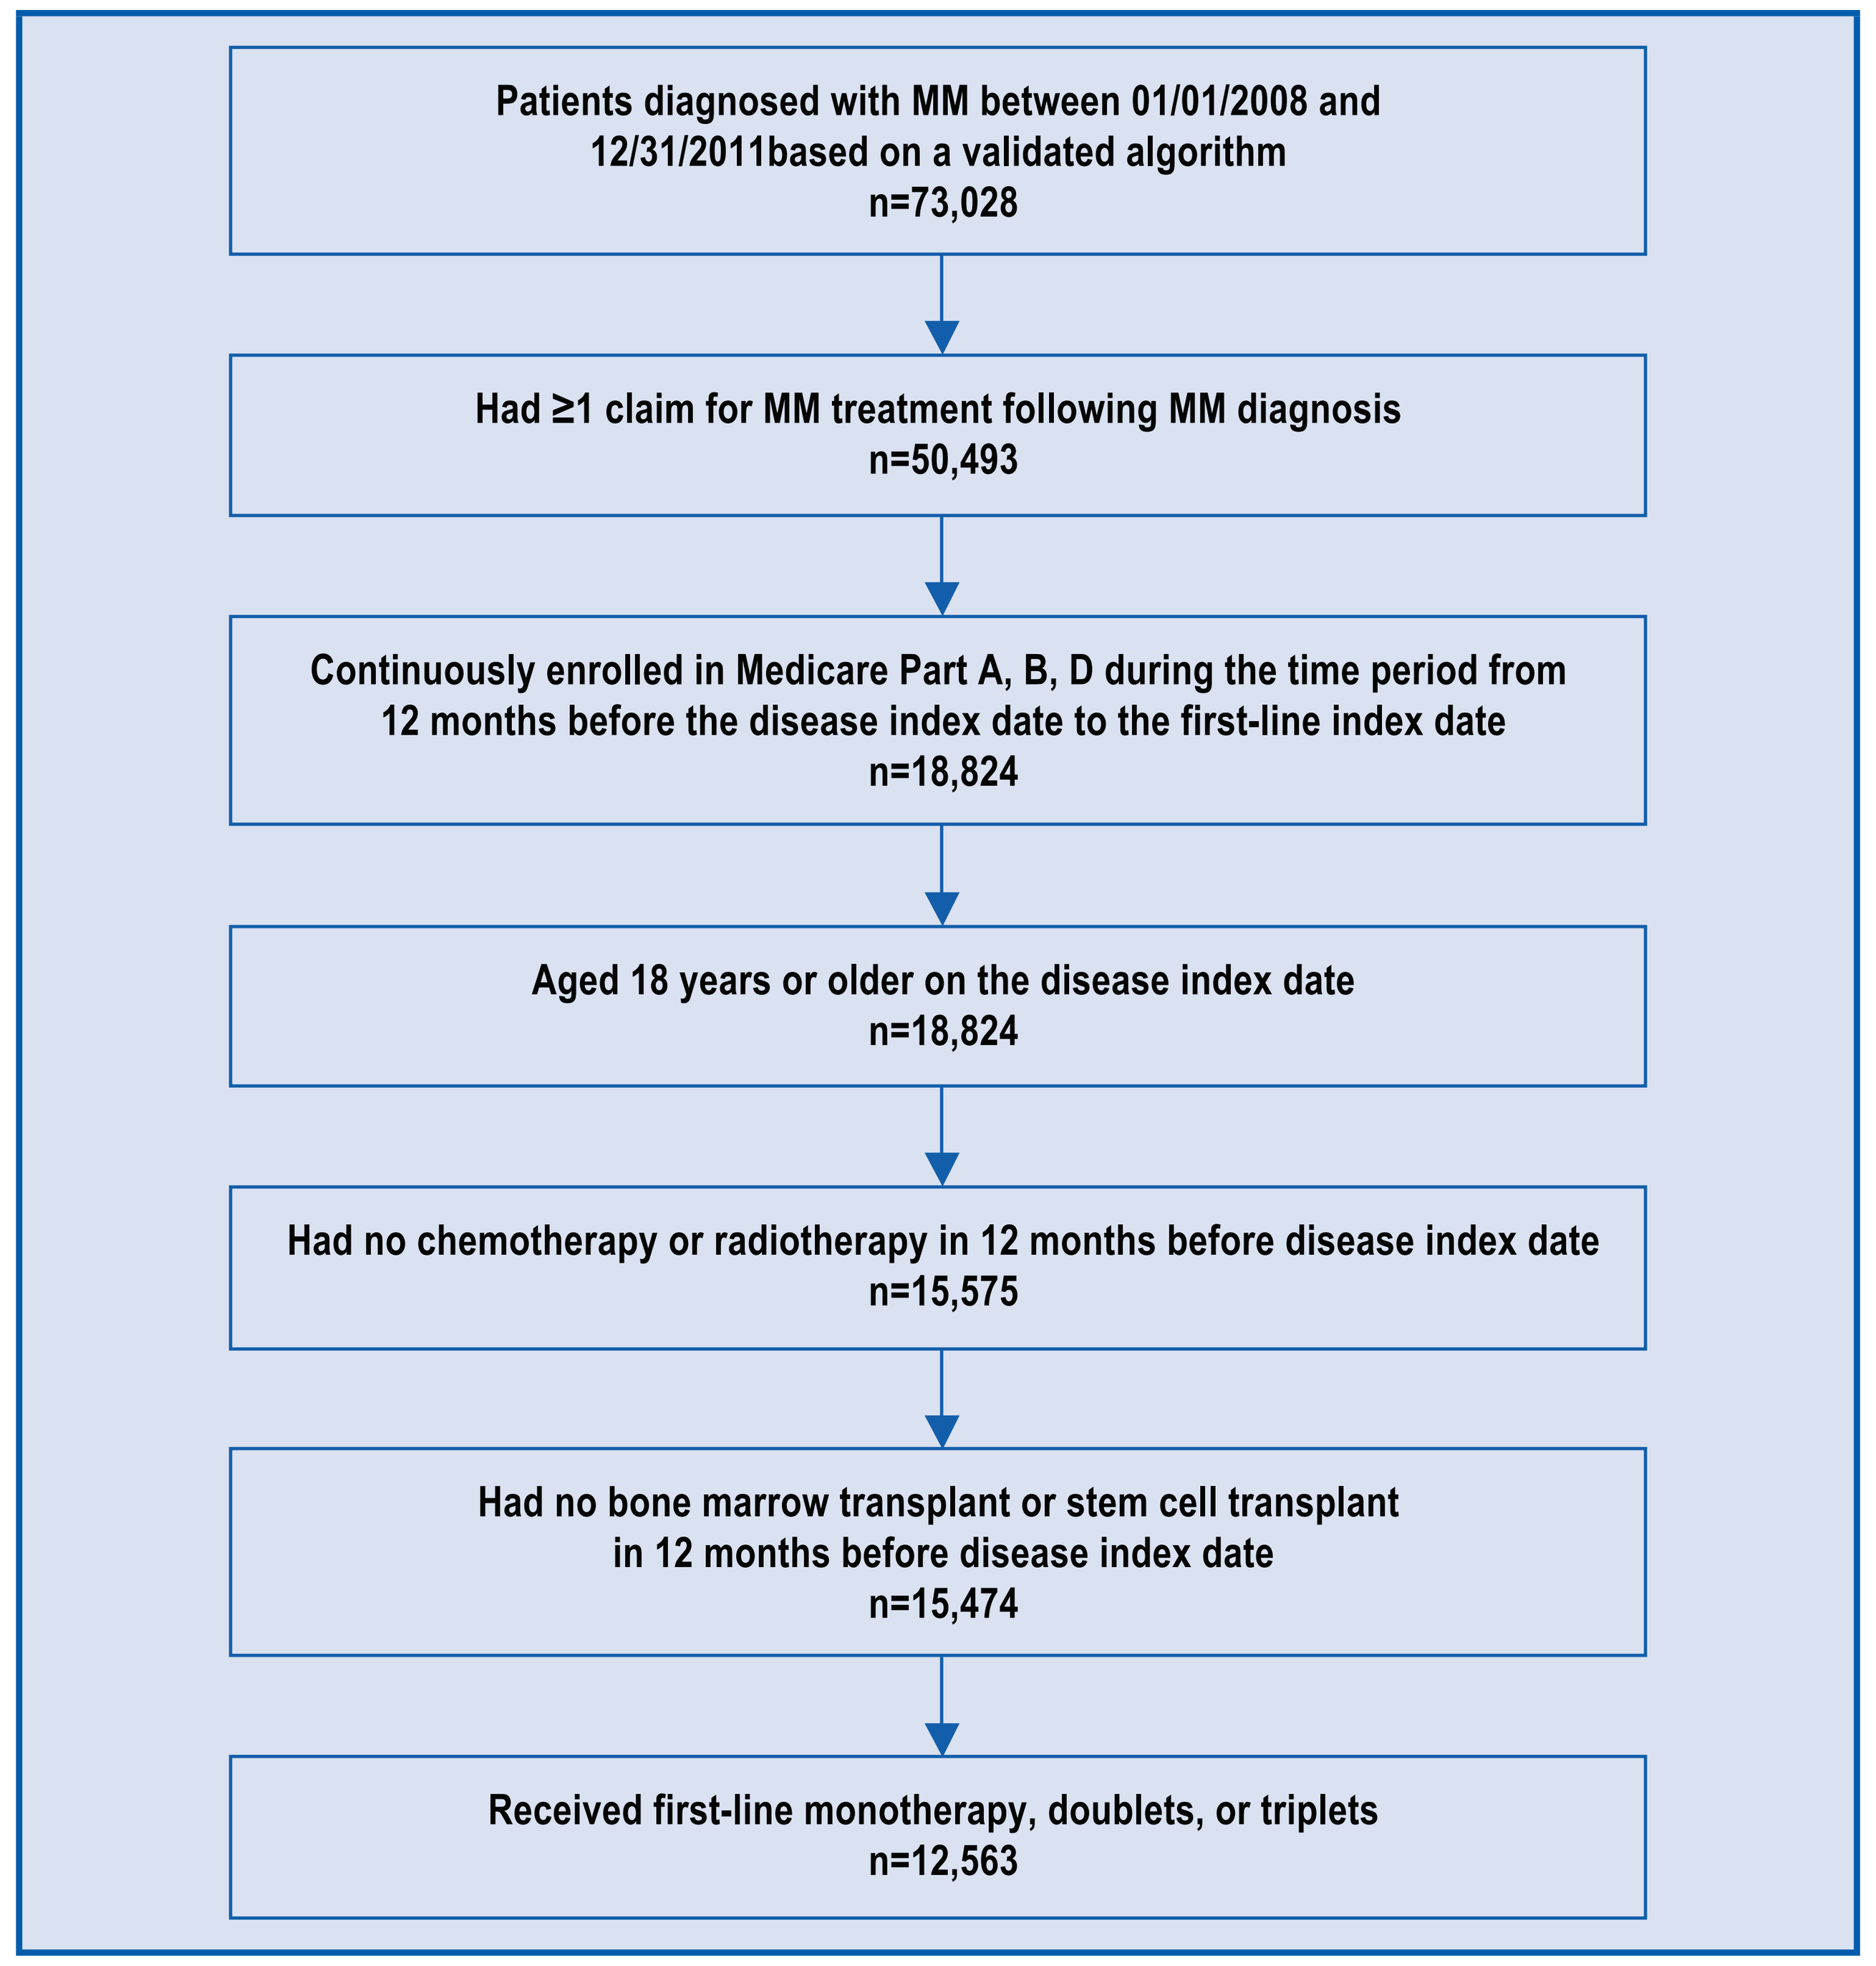

Patients diagnosed with MM between 01/01/2008 and 12/31/2011based on a validated algorithm
n=73,028
Had ≥1 claim for MM treatment following MM diagnosis
n=50,493
Continuously enrolled in Medicare Part A, B, D during the time period from 12 months before the disease index date to the first-line index date
n=18,824
Aged 18 years or older on the disease index date
n=18,824
Had no chemotherapy or radiotherapy in 12 months before disease index date
n=15,575
Had no bone marrow transplant or stem cell transplant in 12 months before disease index date
n=15,474
Received first-line monotherapy, doublets, or triplets
n=12,563

Table 1. Demographic and clinical characteristics of study population by line of therapy

	First-line	Second-line	Third-line	Fourth-line
N	12,563	5647	2243	773
Age mean (SD), years[a]	74.8 (8.5)	73.8 (8.1)	73.0 (7.7)	72.5 (7.5)
Age, %[a]				
18–64[b]	8.5	9.1	9.0	8.9
65–74	42.0	47.1	52.2	55.1
75+	49.5	43.9	38.8	36.0
Sex, %				
Male	46.6	47.5	47.5	45.8
Female	53.4	52.5	52.5	54.2
Race, %				
White	77.6	79.0	81.0	81.1
Black	16.3	14.9	13.4	13.8
Other	6.1	6.1	5.6	5.1
Line index year, %				
2008	20.4	5.7	0.8	*
2009	24.2	18.2	10.6	^
2010	24.9	25.4	23.1	19.7
2011	26.2	26.9	30.5	34.4
2012	4.3	23.8	35.0	42.7
Charlson comorbidty index, %				
0	19.7	1.3	1.2	1.6
1–3	56.5	63.6	66.6	64.3
4+	23.8	35.1	32.3	34.2
Selected comorbid conditions, %				
CHF	16.2	19.1	19.8	24.1
Diabetes	27.5	29.6	30.3	29.0
COPD	16.8	20.3	21.8	24.6
CKD	35.8	41.3	44.9	47.9
Anemia	58.3	72.7	78.8	83.1
Osteoporosis	11.7	14.9	17.4	18.8
Neutropenia	1.4	7.2	14.0	18.9
Thrombocytopenia	6.9	13.6	19.7	23.0
Peripheral neuropathy	6.9	15.9	22.4	26.5

[a]Age defined at disease index date; [b]Enrolled in Medicare due to disabilities or end-stage renal disease
*Values for cells with ten or fewer patients are suppressed
^Value is suppressed to avoid deriving values for other cell with ten or fewer patients
CHF, congestive heart disease; CKD, chronic kidney disease; COPD, chronic obstructive pulmonary disease; SD, standard deviation

- Line of therapy duration and frequency of drug regimens for each line are shown in **Table 2**
 - From line 1 to line 4, line duration decreased overall and by each regimen. Patients treated with monotherapy had the shortest line duration across all 4 lines
 - Overall, doublets were the most common first-line regimen (62%), followed by monotherapy (21%) and triplets (17%). The pattern was similar among patients who advanced to second-, third-, and fourth-line, respectively, though more triplets were used at advanced lines
 - In patients aged ≥75 years, only 12% received triplets in first-line, in contrast to >20% of triplet use in third- and fourth-line, respectively

Table 2. Line of therapy duration and frequency of drug regimens by line of therapy

Therapy	Line of therapy duration in days, mean (SD)	Frequency of regimen, n (%): Overall	Age at disease index date, years: 18–64	65–74	≥75
First-line					
Overall	389 (332)	12,563 (100.0)	1072 (100.0)	5274 (100.0)	6217 (100.0)
Monotherapy	324 (331)	2653 (21.1)	252 (23.5)	921 (17.5)	1480 (23.8)
Doublets	406 (336)	7804 (62.1)	592 (55.2)	3212 (60.9)	4000 (64.3)
Triplets	405 (309)	2106 (16.8)	228 (21.3)	1141 (21.6)	737 (11.9)
Second-line					
Overall	330 (280)	5647 (100.0)	512 (100.0)	2659 (100.0)	2476 (100.0)
Monotherapy	301 (271)	1365 (24.2)	142 (27.7)	624 (23.5)	599 (24.2)
Doublets	346 (287)	3118 (55.2)	263 (51.4)	1427 (53.7)	1428 (57.7)
Triplets	322 (267)	1164 (20.6)	107 (20.9)	608 (22.9)	449 (18.1)
Third-line					
Overall	268 (230)	2243 (100.0)	202 (100.0)	1170 (100.0)	871 (100.0)
Monotherapy	238 (217)	602 (26.8)	68 (33.7)	312 (26.7)	222 (25.5)
Doublets	288 (243)	1131 (50.4)	93 (46.0)	569 (48.6)	469 (53.8)
Triplets	258 (212)	510 (22.7)	41 (20.3)	289 (24.7)	180 (20.7)
Fourth-line					
Overall	233 (203)	773 (100.0)	69 (100.0)	426 (100.0)	278 (100.0)
Monotherapy	199 (208)	197 (25.5)	23 (33.3)	99 (23.2)	75 (27.0)
Doublets	258 (219)	386 (49.9)	32 (46.4)	211 (49.5)	143 (51.4)
Triplets	220 (156)	190 (24.6)	14 (20.3)	116 (27.2)	60 (21.6)

SD, standard deviation

- The top five most common regimens for each line are shown in **Figure 2**
 - Lenalidomide-steroid (Rd), bortezomib-steroid (Vd), bortezomib-lenalidomide-steroid (VRd), and dexamethasone monotherapy were the top four most frequently used regimens across first- to fourth-line
 - The fifth common regimen was thalidomide-steroid (Td) in first-line, lenalidomide monotherapy in second- and third-line, and cyclophosphamide-bortezomib-steroid (CyBorD) in fourth-line

Figure 2. Top five most frequently used drug combinations by line of therapy (%)

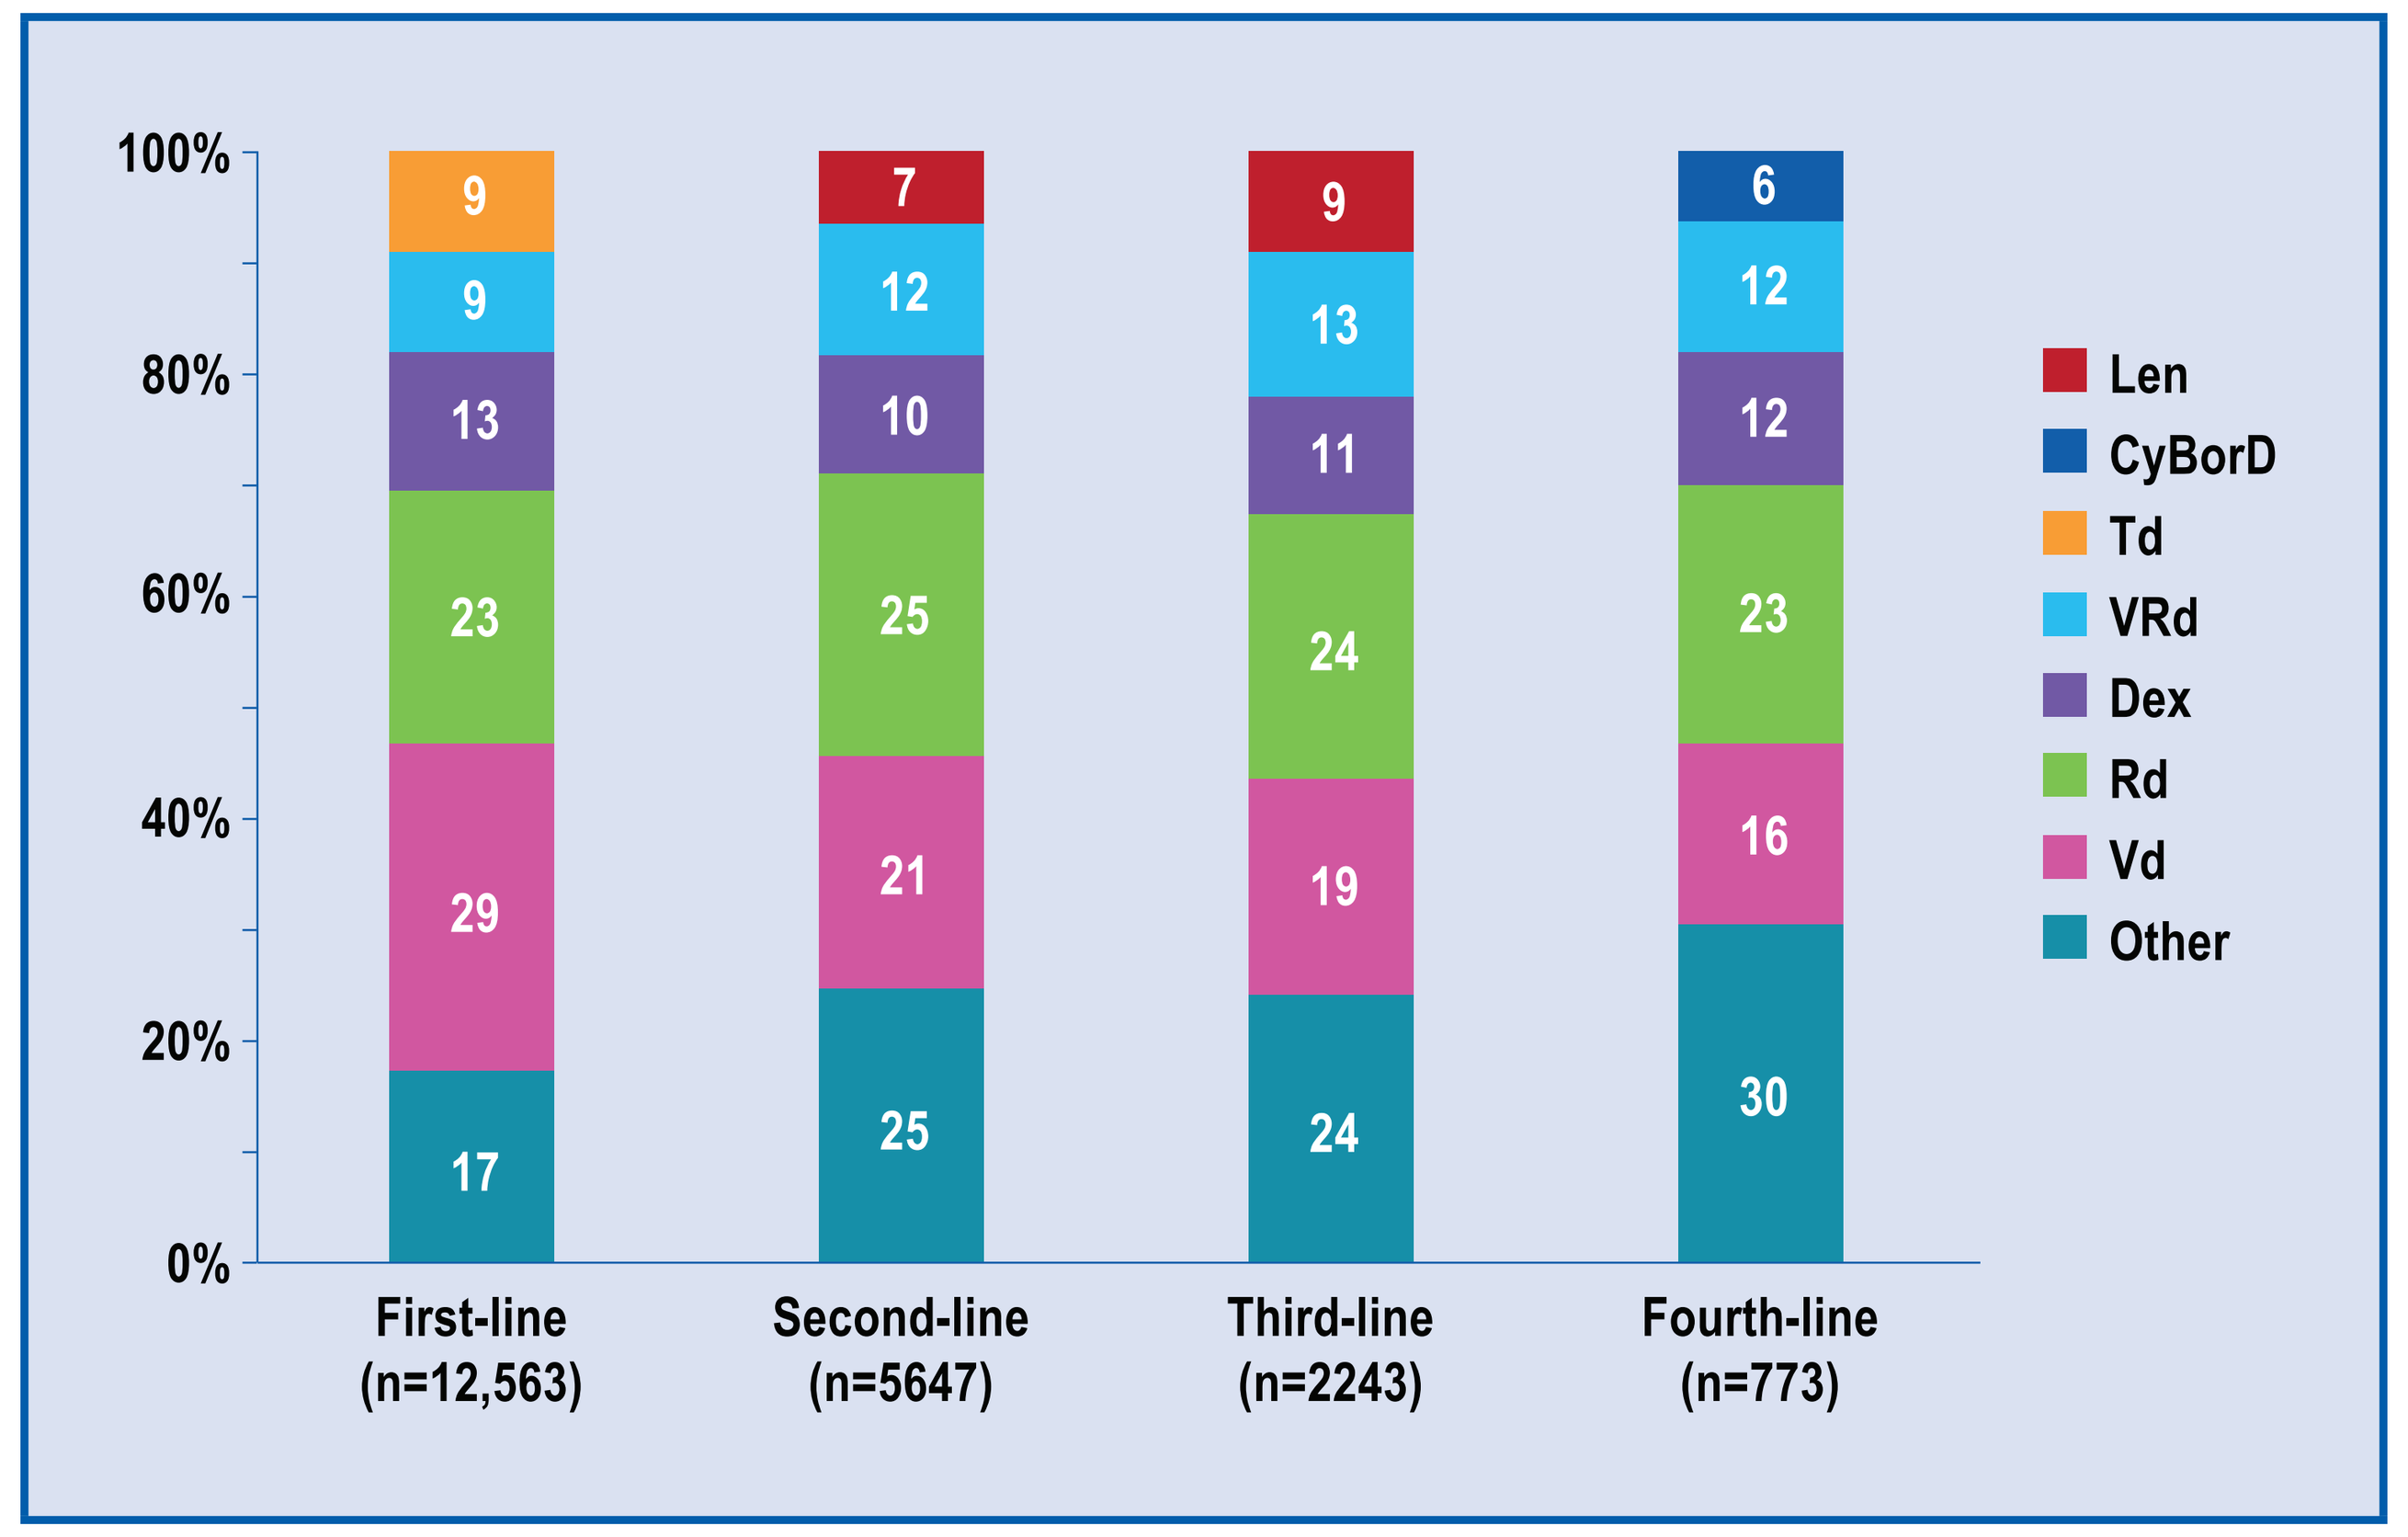

100%
80%
60%
40%
20%
0%
9
9
13
23
29
17
7
12
10
25
21
25
9
13
11
24
19
24
6
12
12
23
16
30
First-line (n=12,563)
Second-line (n=5647)
Third-line (n=2243)
Fourth-line (n=773)
Len
CyBorD
Td
VRd
Dex
Rd
Vd
Other

Dex, dexamethasone; Len, lenalidomide; steroid, dexamethasone or prednisone; Vd, bortezomib-steroid; Rd, lenalidomide-steroid; Td, thalidomide-steroid; VRd, bortezomib-lenalidomide-steroid; CyBorD, cyclophosphamide-bortezomib-steroid

- Treatment sequencing patterns for patients who advanced to second-, third-, and fourth-line are shown in **Figure 3**
 - For patients who advanced to second-line:
 - Of those who received monotherapy in first-line, 37% continued monotherapy and 63% switched to more dense regimens (doublets, 53%; triplets 10%)
 - Of those who received doublets in first-line, 58% continued doublets, 22% switched to triplets, and 20% to monotherapy
 - Of those who received triplets in first-line, 26% continued triplets and 74% switched to less dense regimens (doublets, 47%; monotherapy, 27%)
 - For patients who advanced to third- or fourth-line, the sequencing of prior monotherapy and doublets to current regimens was similar to patients advancing to second-line; however, sequencing of triplet to triplet increased to 32% and 37% of third- and fourth-line patients who received prior triplets, respectively

Figure 3. Treatment sequencing patterns for patients who advanced to second-, third-, and fourth-line by type of drug regimens in the prior line of therapy

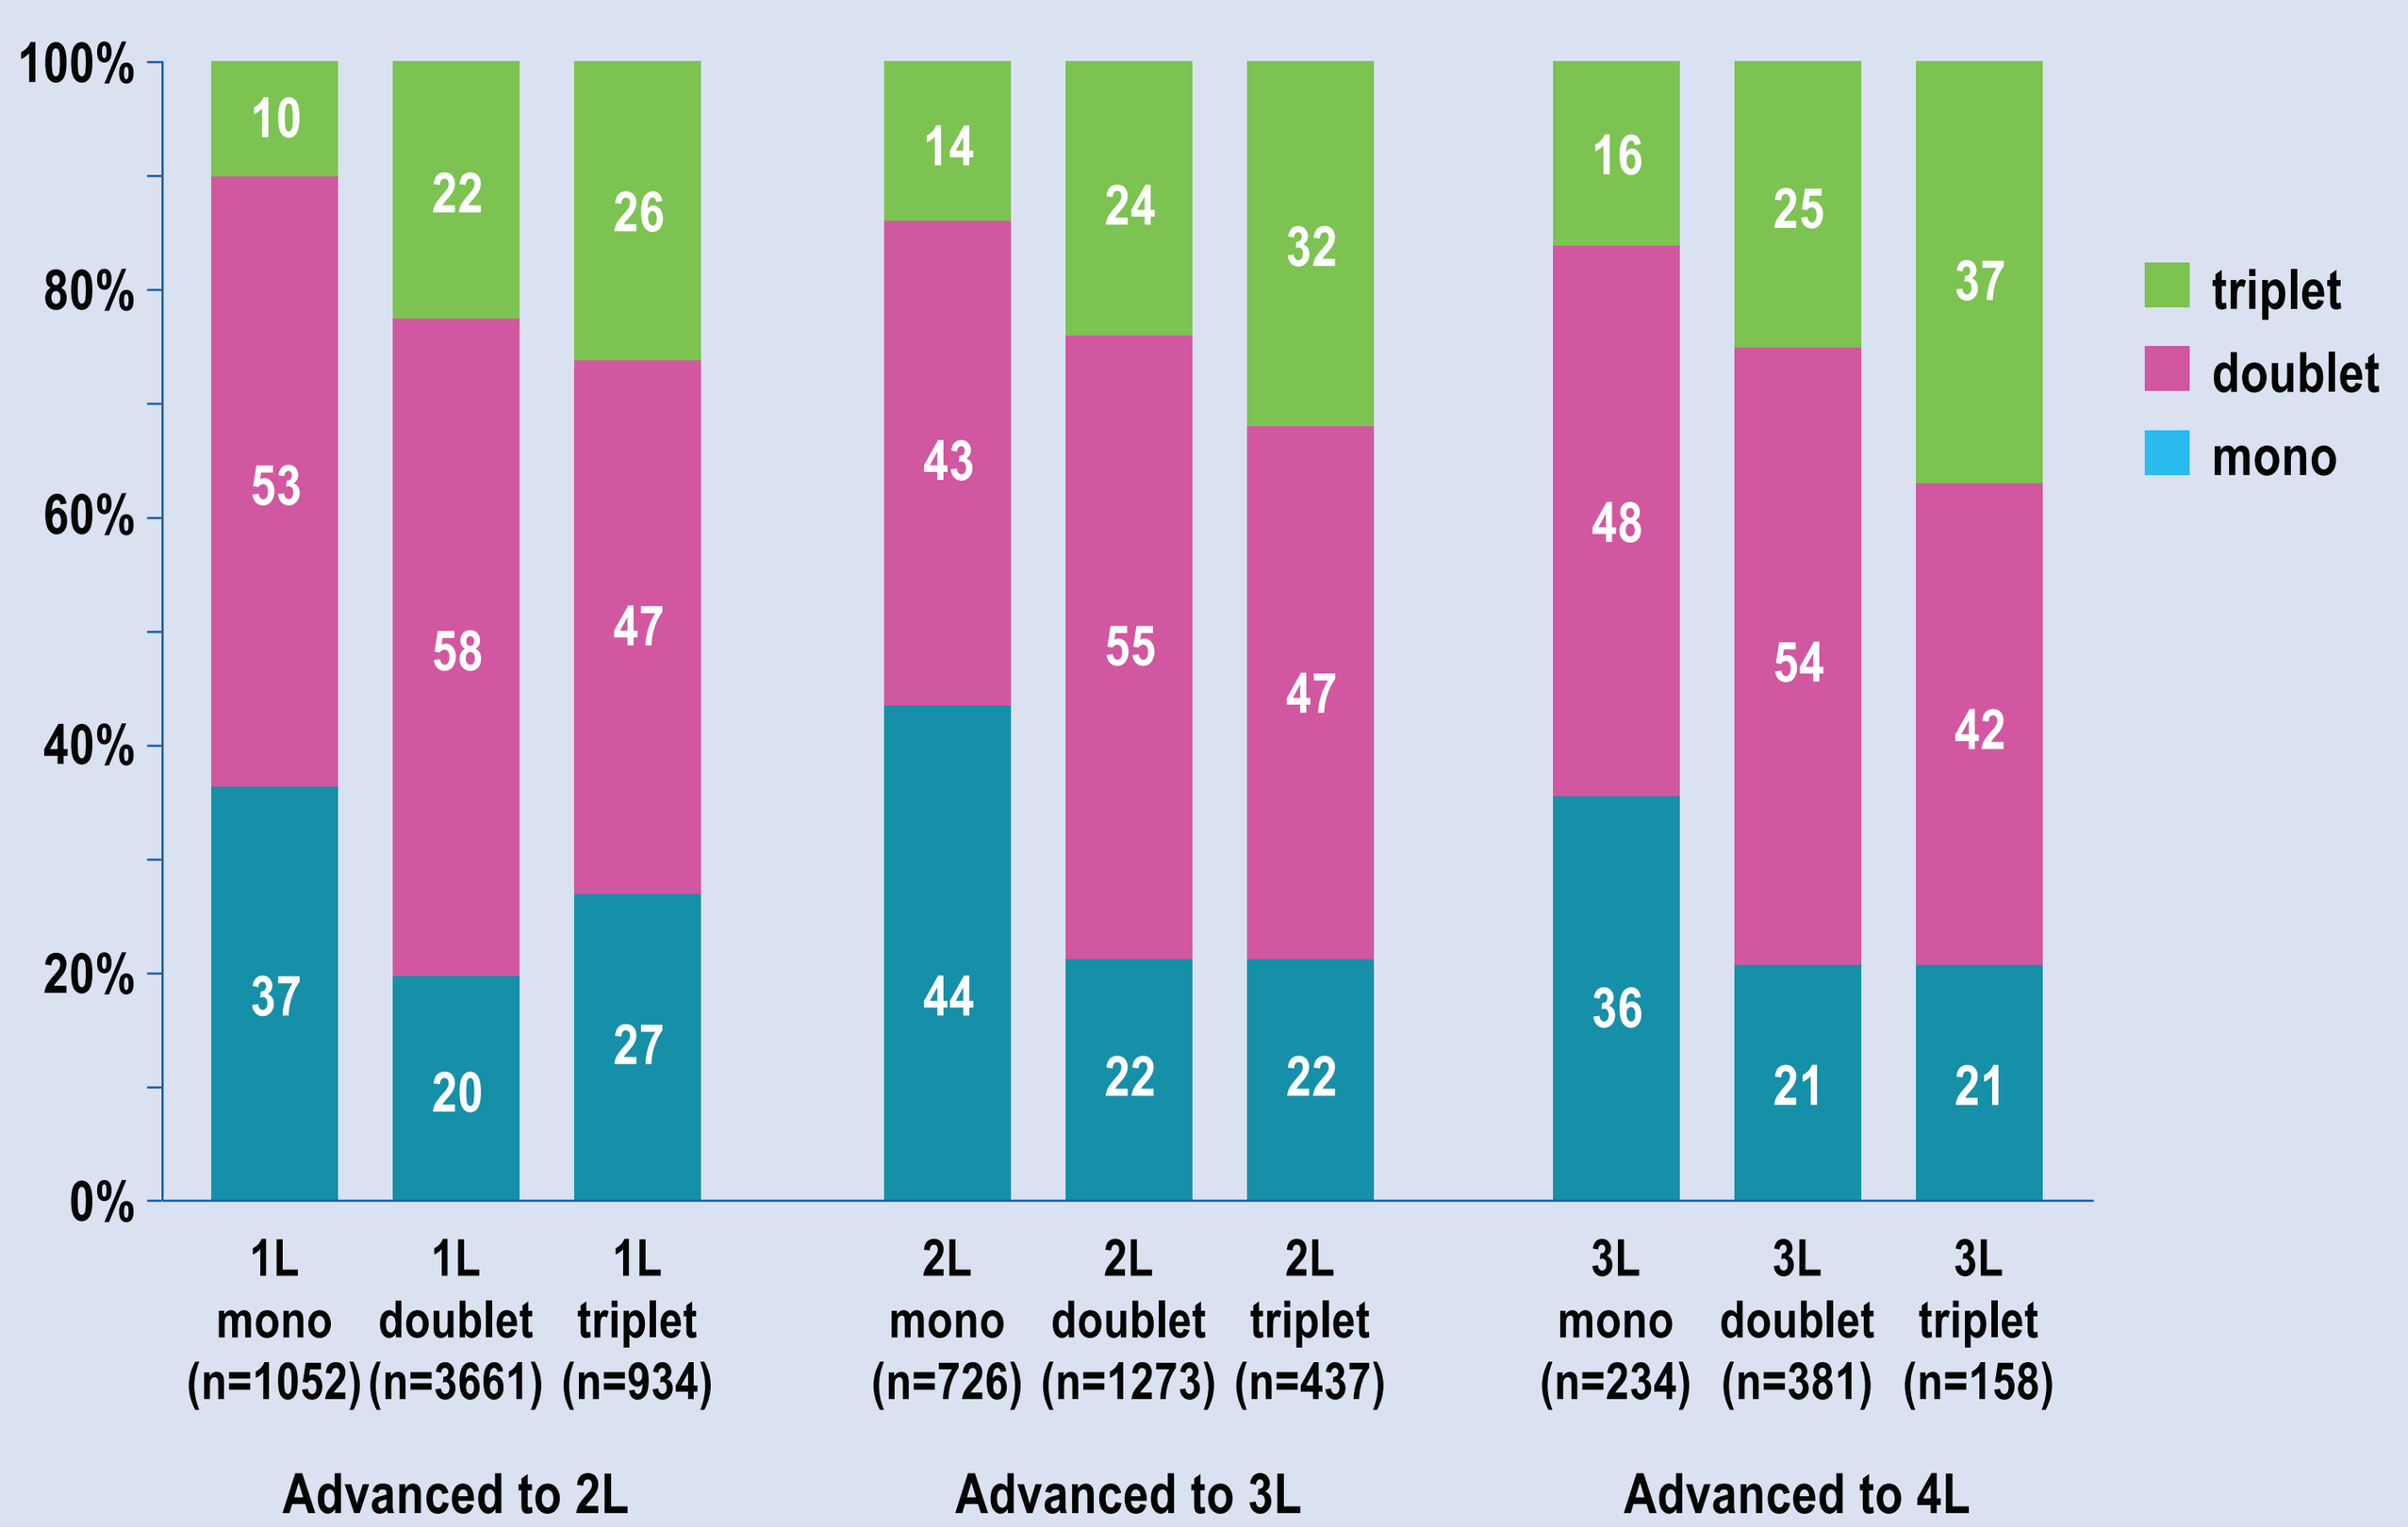

100%
80%
60%
40%
20%
0%
10
53
37
22
58
20
26
47
27
14
43
44
24
55
22
32
47
22
16
48
36
25
54
21
37
42
21
1L mono (n=1052)
1L doublet (n=3661)
1L triplet (n=934)
2L mono (n=726)
2L doublet (n=1273)
2L triplet (n=437)
3L mono (n=234)
3L doublet (n=381)
3L triplet (n=158)
Advanced to 2L
Advanced to 3L
Advanced to 4L
triplet
doublet
mono

1L, First-line; 2L, Second-line; 3L, Third-line; 4L, Fourth-line; mono, Monotherapy

CONCLUSIONS

- **Among Medicare patients with MM, doublets were the most frequently used regimens across all lines of therapy, while triplets were used in more advanced lines**
- **Patients on monotherapy or doublets were more likely to retain their treatment pattern when they advance to the next line of therapy, while those on triplet regimen were more likely to switch to a less dense regimen when they advance to their next line of therapy**
- **Fewer patients of older age (≥75 years) were prescribed triplet therapies; however, triplet use in this patient group increases in more advanced lines. This may reflect increasing community awareness of emerging data on the superior efficacy of triplets compared to doublets regimens**
- **These results provide a baseline description of treatment patterns from which we will be able to benchmark the impact of the recent introduction of novel agents and their use in elderly MM patients**
- **Further studies assessing the comparative effectiveness and benefit-risk of treatment sequences are warranted**

REFERENCES

1. Kumar et al. Blood 2008;111:2516–20
2. NCCN Clinical Practice Guidelines in Oncology. Multiple Myeloma Version 2.2014
3. Song et al. Curr Med Res Opin 2016;32(1):95–103
4. Princic et al. Blood 2015;126:4521

ACKNOWLEDGMENTS

This study was supported by Amgen, Inc. The authors thank BlueMomentum, an Ashfield Company, part of UDG Healthcare PLC, for poster editing funded by Amgen, Inc.

CONFLICTS OF INTEREST

Dr. Li and Ms. Natwick report no conflicts of interest. Drs. Yusuf, Vidito, Mezzi, and Werther are employees of Amgen, Inc.

Copies of this poster obtained through Quick Response Code are for personal use only and may not be reproduced without permission from ASH® and the author of this poster

© 2016 Amgen Inc.

American Society of Hematology (ASH) annual meeting; December 3–6, 2016, in San Diego, CA, USA